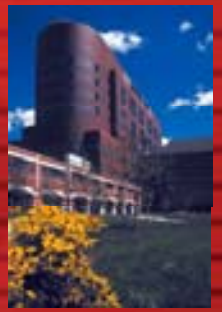

Serum endoglin (CD105): Molecular heterogeneity and marked differences among different anti-endoglin monoclonal antibodies in the detection of metastasis and tumor progression

Naoko Harada, Norihiro Takahashi, Yuro Haruta, Hilda Tsai, Jill Duzen, Kanako Fujita, William Greco, Ben K. Seon, Roswell Park Cancer Institute, Buffalo, NY

Abstract

Endoglin (ENG, CD105) is a proliferation-associated homodimeric cell membrane antigen of endothelial cells and leukemia cells. It is strongly expressed on angiogenic vascular endothelium in tumors and also expressed on lymphatic vessels in tumors. Furthermore, ENG is a TGF-β coreceptor and essential for angiogenesis. ENG represents a more specific marker for tumor angiogenesis and/or tumor progression than the commonly used pan-endothelial markers such as CD34 and CD31.

Previously we developed a double antibody sandwich radioimmunoassay (DAS-RIA) using two anti-ENG monoclonal antibodies (mAbs) SN6h and SN6a to quantify nanograms/ml levels of serum ENG and found association of serum ENG levels with tumor metastasis (Clin Cancer Res, 2001, 7: 524). The objective of the present study is to characterize molecular nature of serum ENG and to test a hypothesis that sensitivity and specificity of DAS-RIA can be substantially improved by selecting appropriated pairs of anti-ENG mAbs. We generated 12 anti-ENG mAbs and epitopes defined by these mAbs were mapped. We selected 5 pairs of anti-ENG mAbs defining different epitopes to compare their sensitivity and specificity. Anti-ENG mAb SN6h was used as the capture antibody in each pair because SN6h possesses an extremely high antigen-binding avidity (association K = 1.38 x 1011 liters/mol) and it strongly binds both native and denatured forms of ENG. The order of sensitivity among the five pairs was determined to be SN6f/SN6h>> SN6g/SN6h> SN6a/SN6h ≈ SN6j/SN6h> SN6/SN6h. SN6f/SN6h and SN6g/SN6h pairs were particularly effective for distinguishing between Metastasis-positive (Meta+) and Meta- samples. In an additional study, we developed a sensitive DAS-ELISA using SN6f/SN6h and SN6g/SN6h to facilitate easier quantification of soluble ENG. Reasons for the different capacity of different pairs of anti-ENG mAbs to distinguish between Meta+ and Meta- samples will be attributable to molecular heterogeneity of serum ENG, different epitopes defined by individual mAbs, and differences in the antigen-binding avidity among these mAbs. Western blot of serum samples from cancer patients showed highly heterogeneous patterns of serum ENG. Furthermore, certain mAbs were able to detect only a fraction of the heterogeneous ENG molecules while others were capable of detecting a series of heterogeneous ENG molecules. Test results suggest that the major ENG component in the heterogeneous ENG corresponds to a complex of TGF-β1/TGF-β3, TGF-β receptor II and ENG fragment(s). Assay results of serum ENG will be strongly influenced by the molecular heterogeneity of serum ENG, different capacity of individual anti-ENG mAbs in detection of different components of serum ENG, and differences in the heterogeneous patterns of serum ENG among different cancer patients.

Endoglin (CD105)

1. Endoglin (ENG) is a homodimeric cell membrane glycoprotein of leukemia cells and endothelial cells (Haruta and Seon, Proc. Natl. Acad. Sci. 1986; Gougos and Letarte, J. Immunol. 1988).
2. ENG is a proliferation-associated antigen and strongly expressed in angiogenic vascular endothelium of tumor tissues.
3. ENG is expressed in lymphatic endothelium of tumor tissues but was not detectable in lymphatic endothelium of normal tissues.
4. ENG is a TGF- β coreceptor and binds TGF- β specifically.
5. ENG is essential for angiogenesis.

Methodology

- We developed a double-antibody sandwich RIA and ELISA to quantify soluble ENG in the serum/plasma of cancer patients and control subjects.
- Five different pairs of anti-ENG mAbs were compared in the assay to investigate different capacity and sensitivity of the different pairs in distinguishing Meta+ and Meta- serum samples.
- Molecular nature of the soluble ENG as well as other parameters of the mAbs were examined to understand the reasons for the wide variation in the capacity and sensitivity of the different pairs in the assay.

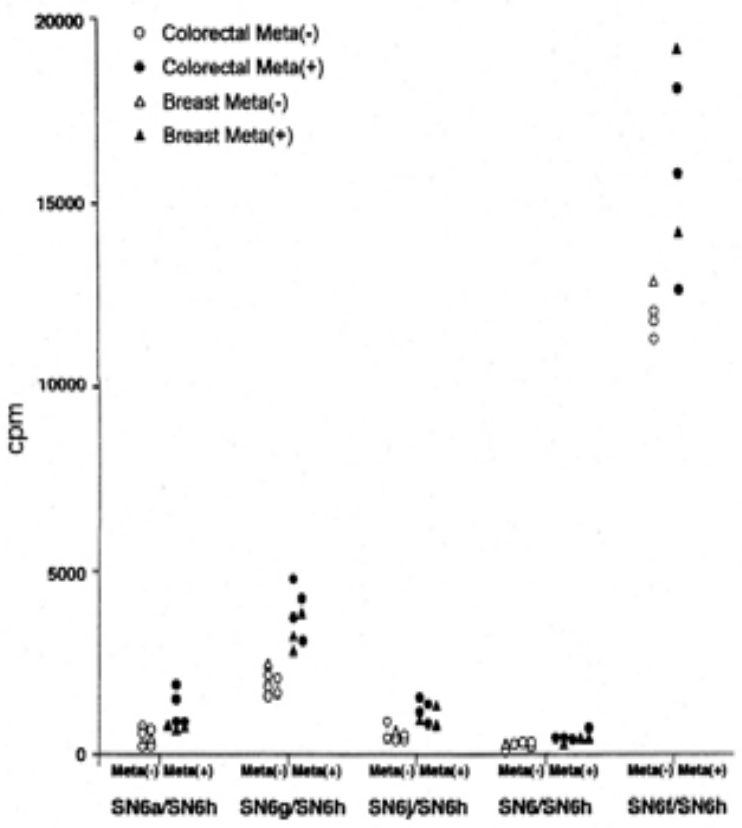
Colorectal Meta(-)
Colorectal Meta(+)
Breast Meta(-)
Breast Meta(+)
cpm
20000
15000
10000
5000
0
Meta(-) Meta(+)
SN6a/SN6h
SN6g/SN6h
SN6j/SN6h
SN6/SN6h
SN6f/SN6h

Different Sensitivity and Efficacy Associated with Different Pairs of Anti-Endoglin mAbs in Double-Antibody Sandwich RIA for Serum Endoglin

Linear (A) and Conformational (B) Epitope Maps of SN6 Series mAbs

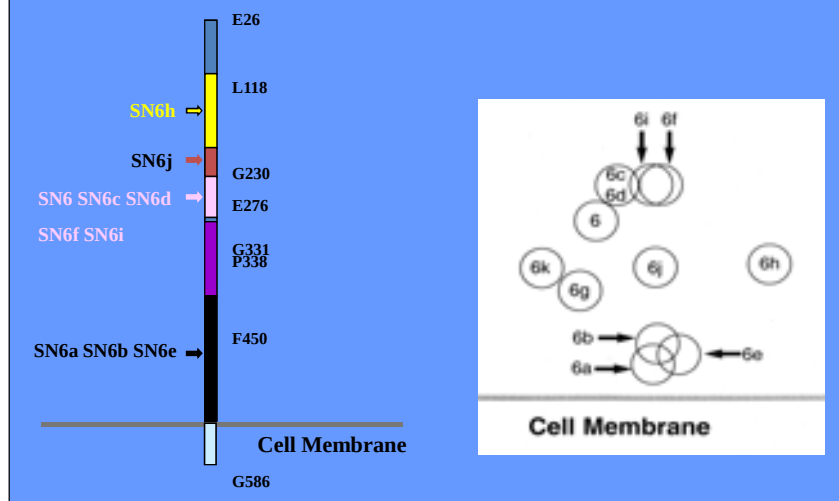
E26
L118
SN6h
SN6j
G230
SN6 SN6c SN6d
E276
SN6f SN6i
G331
P338
F450
SN6a SN6b SN6e
Cell Membrane
G586
6i 6f
6c
6d
6
6k
6g
6j
6h
6b
6a
6e
Cell Membrane

A: Linear Sequence Epitope Map

B: Conformational Epitope Map

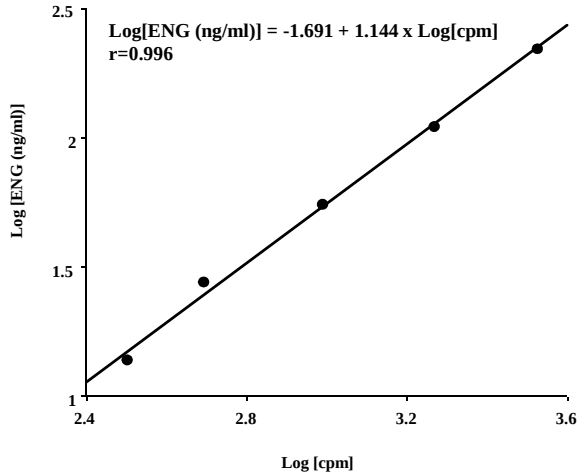
Log[ENG (ng/ml)] = -1.691 + 1.144 x Log[cpm]
r=0.996
Log [ENG (ng/ml)]
Log [cpm]

Standard Curve for Endoglin in a Double-Abs Sandwich RIA

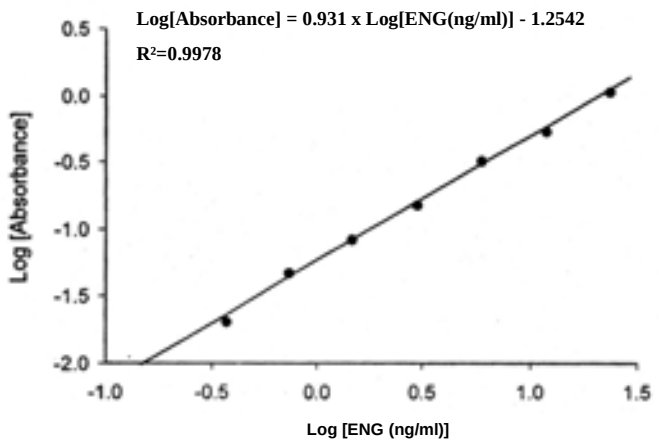
Log[Absorbance] = 0.931 x Log[ENG(ng/ml)] - 1.2542
R²=0.9978
Log [Absorbance]
Log [ENG (ng/ml)]

Standard Curve for Soluble Endoglin as Measured by Double-Antibody (SN6h/SN6g) Sandwich ELISA

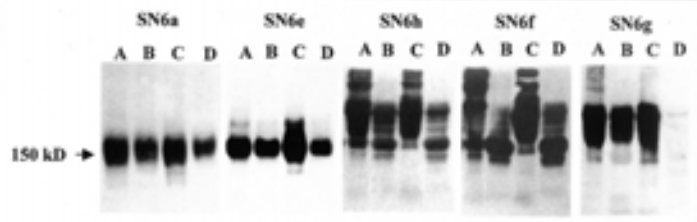
SN6a
SN6e
SN6h
SN6f
SN6g
A B C D
150 kD

Differences/similarity among five anti-ENG mAbs in the capacity to detect heterogeneous ENG components in the serum specimens from two breast (A, B) and two colorectal (C, D) cancer patients

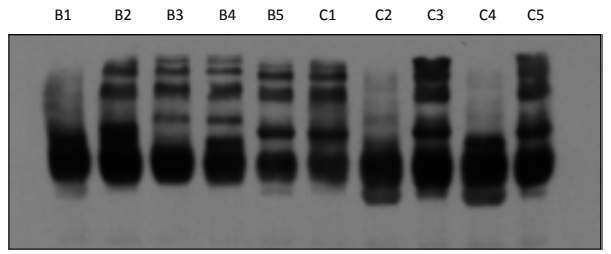
B1 B2 B3 B4 B5 C1 C2 C3 C4 C5

Multiple ENG components in the serum samples from five breast (B1-B5) and five colon (C1-C5) cancer patients

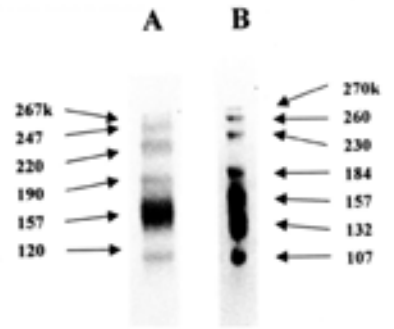
A
B
267k
247
220
190
157
120
270k
260
230
184
157
132
107

Multiple Endoglin Components in the Serum of Metastasis-positive Breast Cancer Patient as Detected by Western Blot/SN6h Using Mini Gels (A) or Regular Size Gels (B)

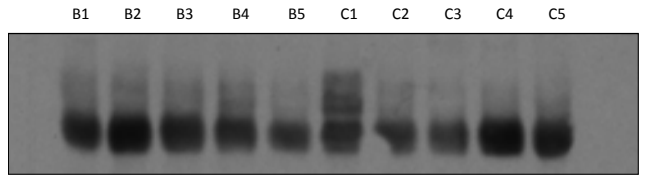
B1 B2 B3 B4 B5 C1 C2 C3 C4 C5

Detection of TGF-β1 in the major component of soluble ENG in the serum samples from five breast (B1-B5) and five colon (C1-C5) cancer patients

Conclusions

- Five pairs of anti-ENG mAbs in the double-antibody sandwich assay showed individually different capacity and sensitivity in distinguishing between metastasis-positive (Meta+) and Meta- serum samples.
- The major causes for the observed differences are attributable to the molecular heterogeneity of soluble ENG, different epitopes defined by individual anti-ENG mAbs, and different antigen binding avidities of the mAbs.